Disruption and Adaptation in Inflammatory Bowel Disease: Impact of the COVID-19 Pandemic on Treatment Patterns, Healthcare Utilization, and Vaccination in the United States

Authors: Komal Kumar, PharmD/MBA Candidate 2026, Scott M. Myers, MBA; Pamala A. Pawloski, PharmD, BCOP, FCCP; Cate Lockhart, PharmD, PhD

BACKGROUND

- The COVID-19 pandemic disrupted IBD care through treatment delays, reduced procedures, and vaccine challenges in immunosuppressed patients.[1–4]
- Healthcare systems rapidly expanded telemedicine to sustain care continuity.[2,5]
- Most immunosuppressive therapies can be continued, as they do not increase severe COVID-19 risk, while corticosteroids should be minimized.[1,2,5]

OBJECTIVES

- Describe national trends in inflammatory bowel disease (IBD) management and treatment patterns.
- This study assessed the impact of COVID-19 on IBD care across four key phases: pre-pandemic (Jan–Dec 2019), early disruption (Mar–May 2020), adaptation (Jun–Dec 2020), and recovery (Jan 2021–Jan 2022).
- Evaluate shifts in care delivery and treatment access, including changes in telehealth utilization, therapy use, vaccination uptake, and hospitalization rates.
- Characterize how pandemic-related disruptions influenced continuity of care and system-level recovery in IBD management.

Change in IBD Encounters During Early COVID-19 (Mar–May 2020 vs 2019)

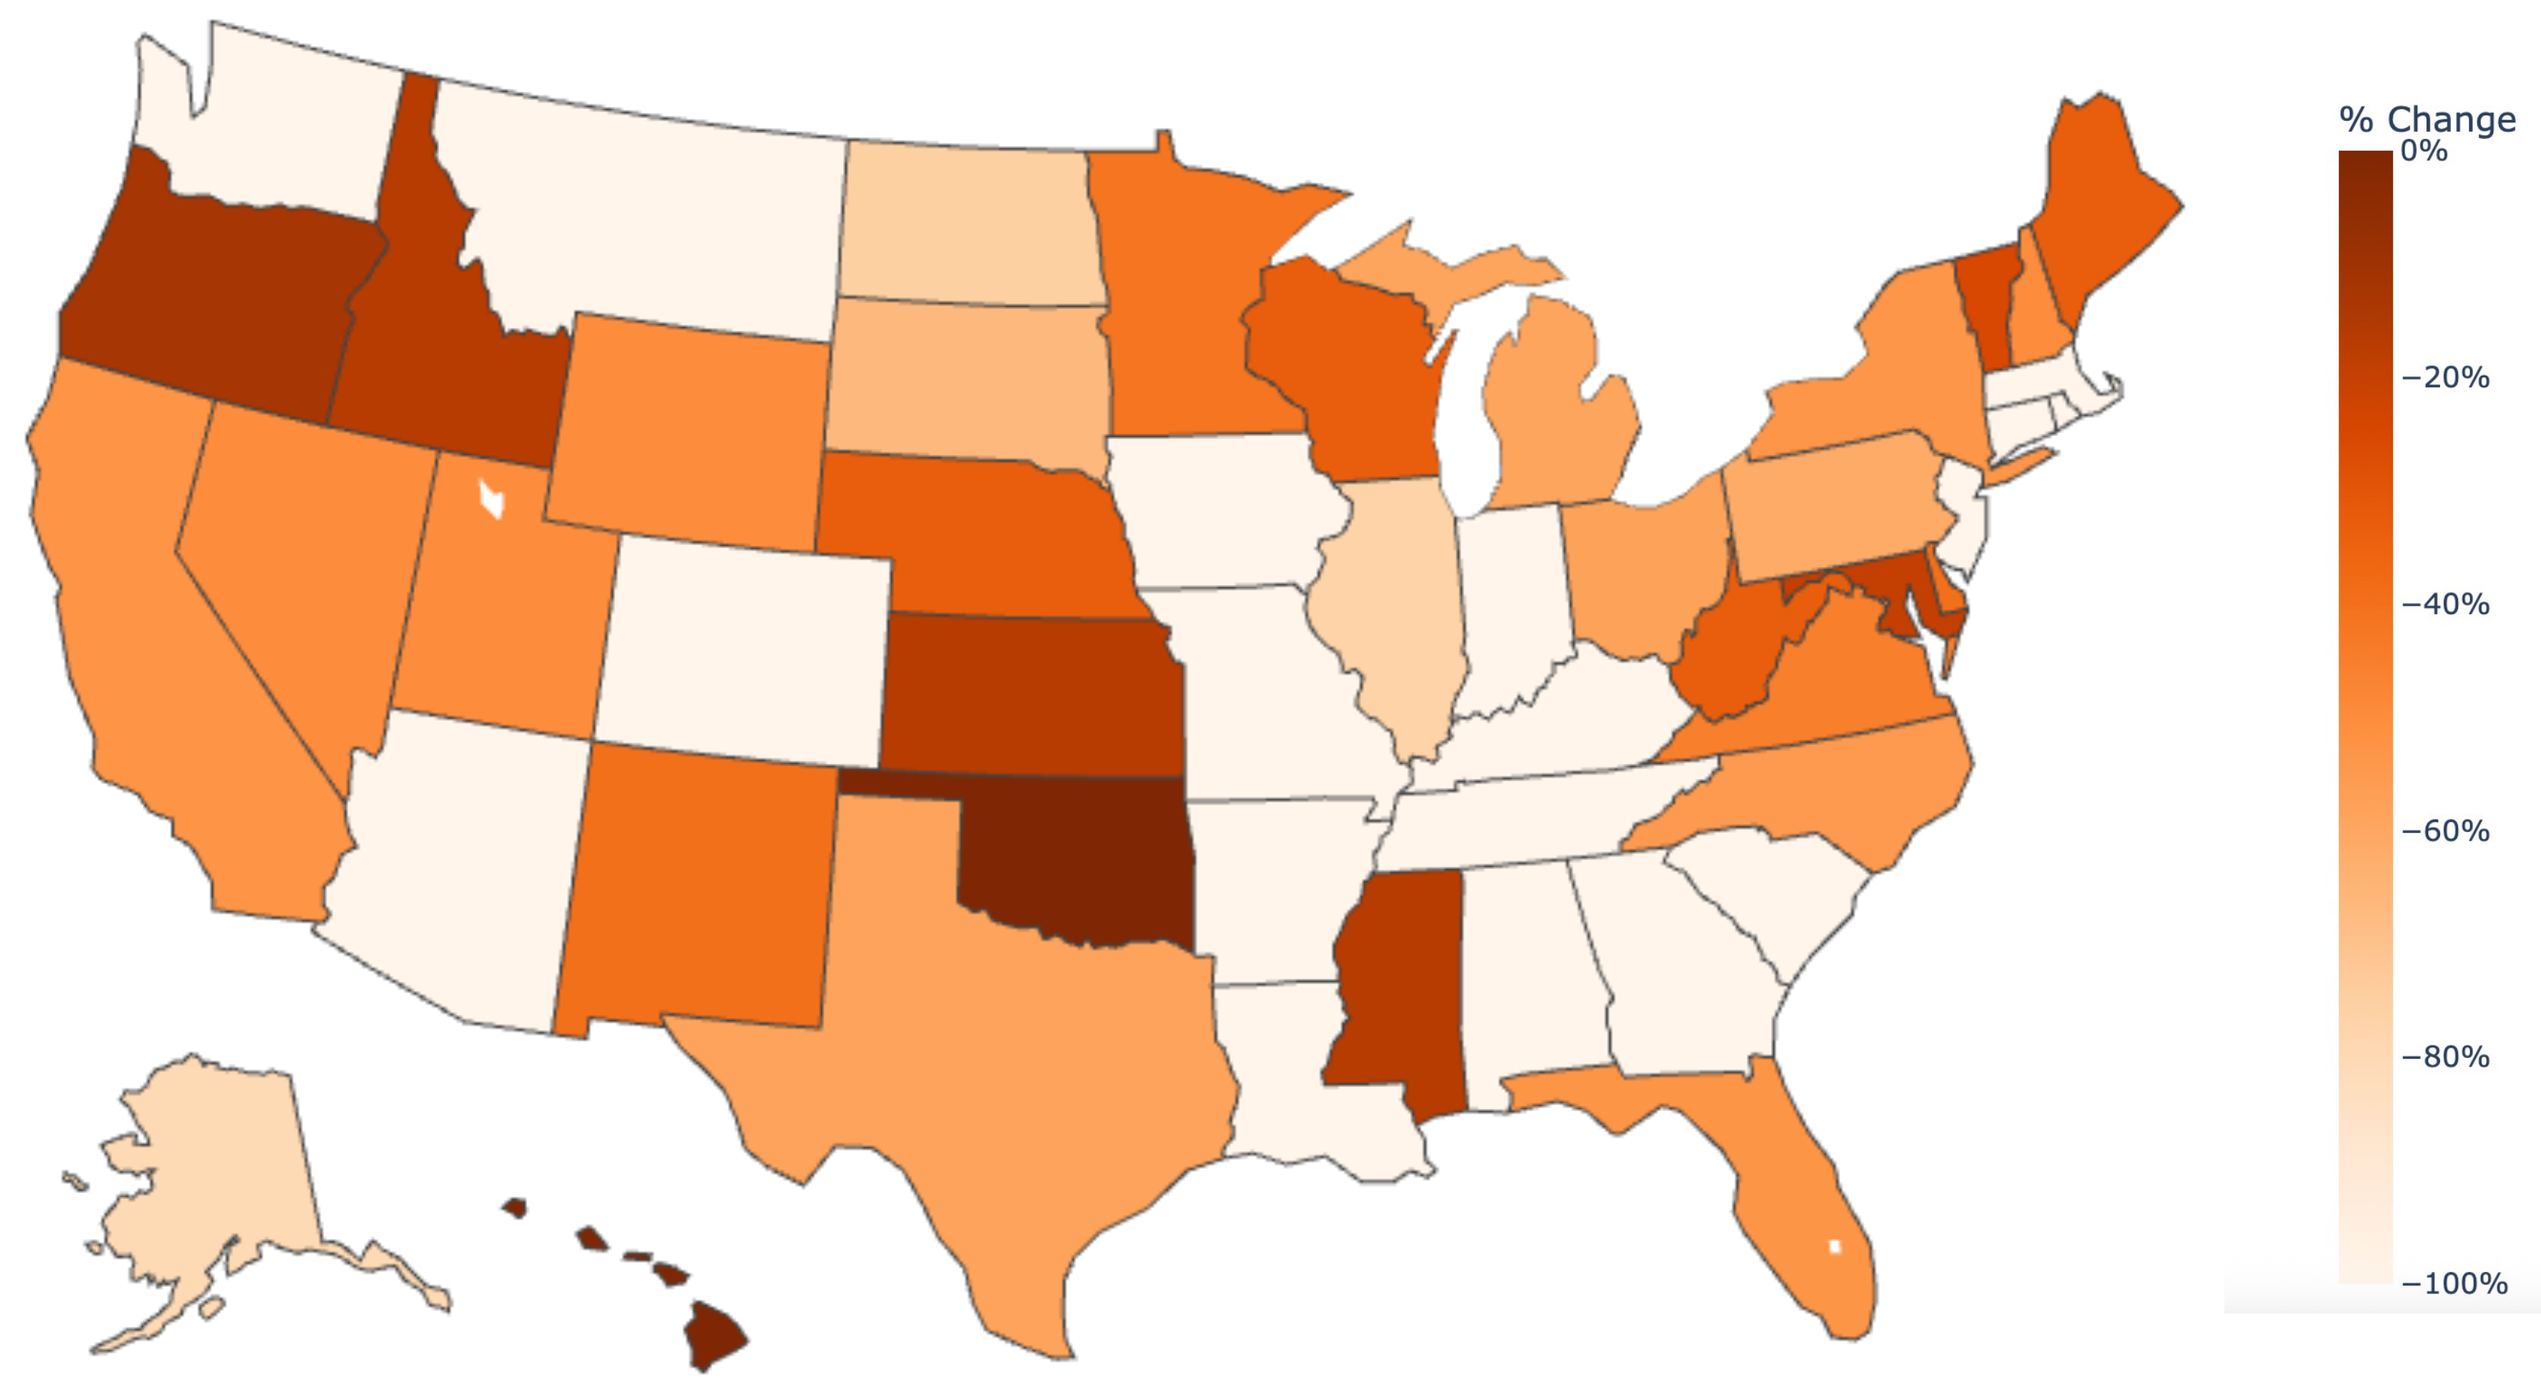
% Change
0%
−20%
−40%
−60%
−80%
−100%

Figure 1. State-Level Change in IBD Encounters During the Early COVID-19 Period (Mar–May 2020 vs 2019).
This U.S. heat map depicts the percent change in inflammatory bowel disease (IBD)–related encounters by state, based on aggregated PearlDiver Mariner claims encompassing office, outpatient, and inpatient visits. Darker orange indicates larger reductions in encounter volume (i.e., greater disruption), whereas lighter shades represent smaller decreases. States shown in gray lacked sufficient baseline data for comparison.

RESULTS

Therapy Classification

Advanced (Biologic / Targeted)
Infliximab, Vedolizumab, Adalimumab, Ustekinumab, Tofacitinib, Baricitinib
Route(s): IV / SC / Oral

Conventional (Immunomodulator / 5-ASA / Corticosteroid)
Azathioprine, Mercaptopurine, Mesalamine, Prednisone
Route(s): Oral / IV

Therapy classification used for analysis based on AGA and ACG guidance.

Monthly IBD Therapy Claims Per 10,000 IBD Patients

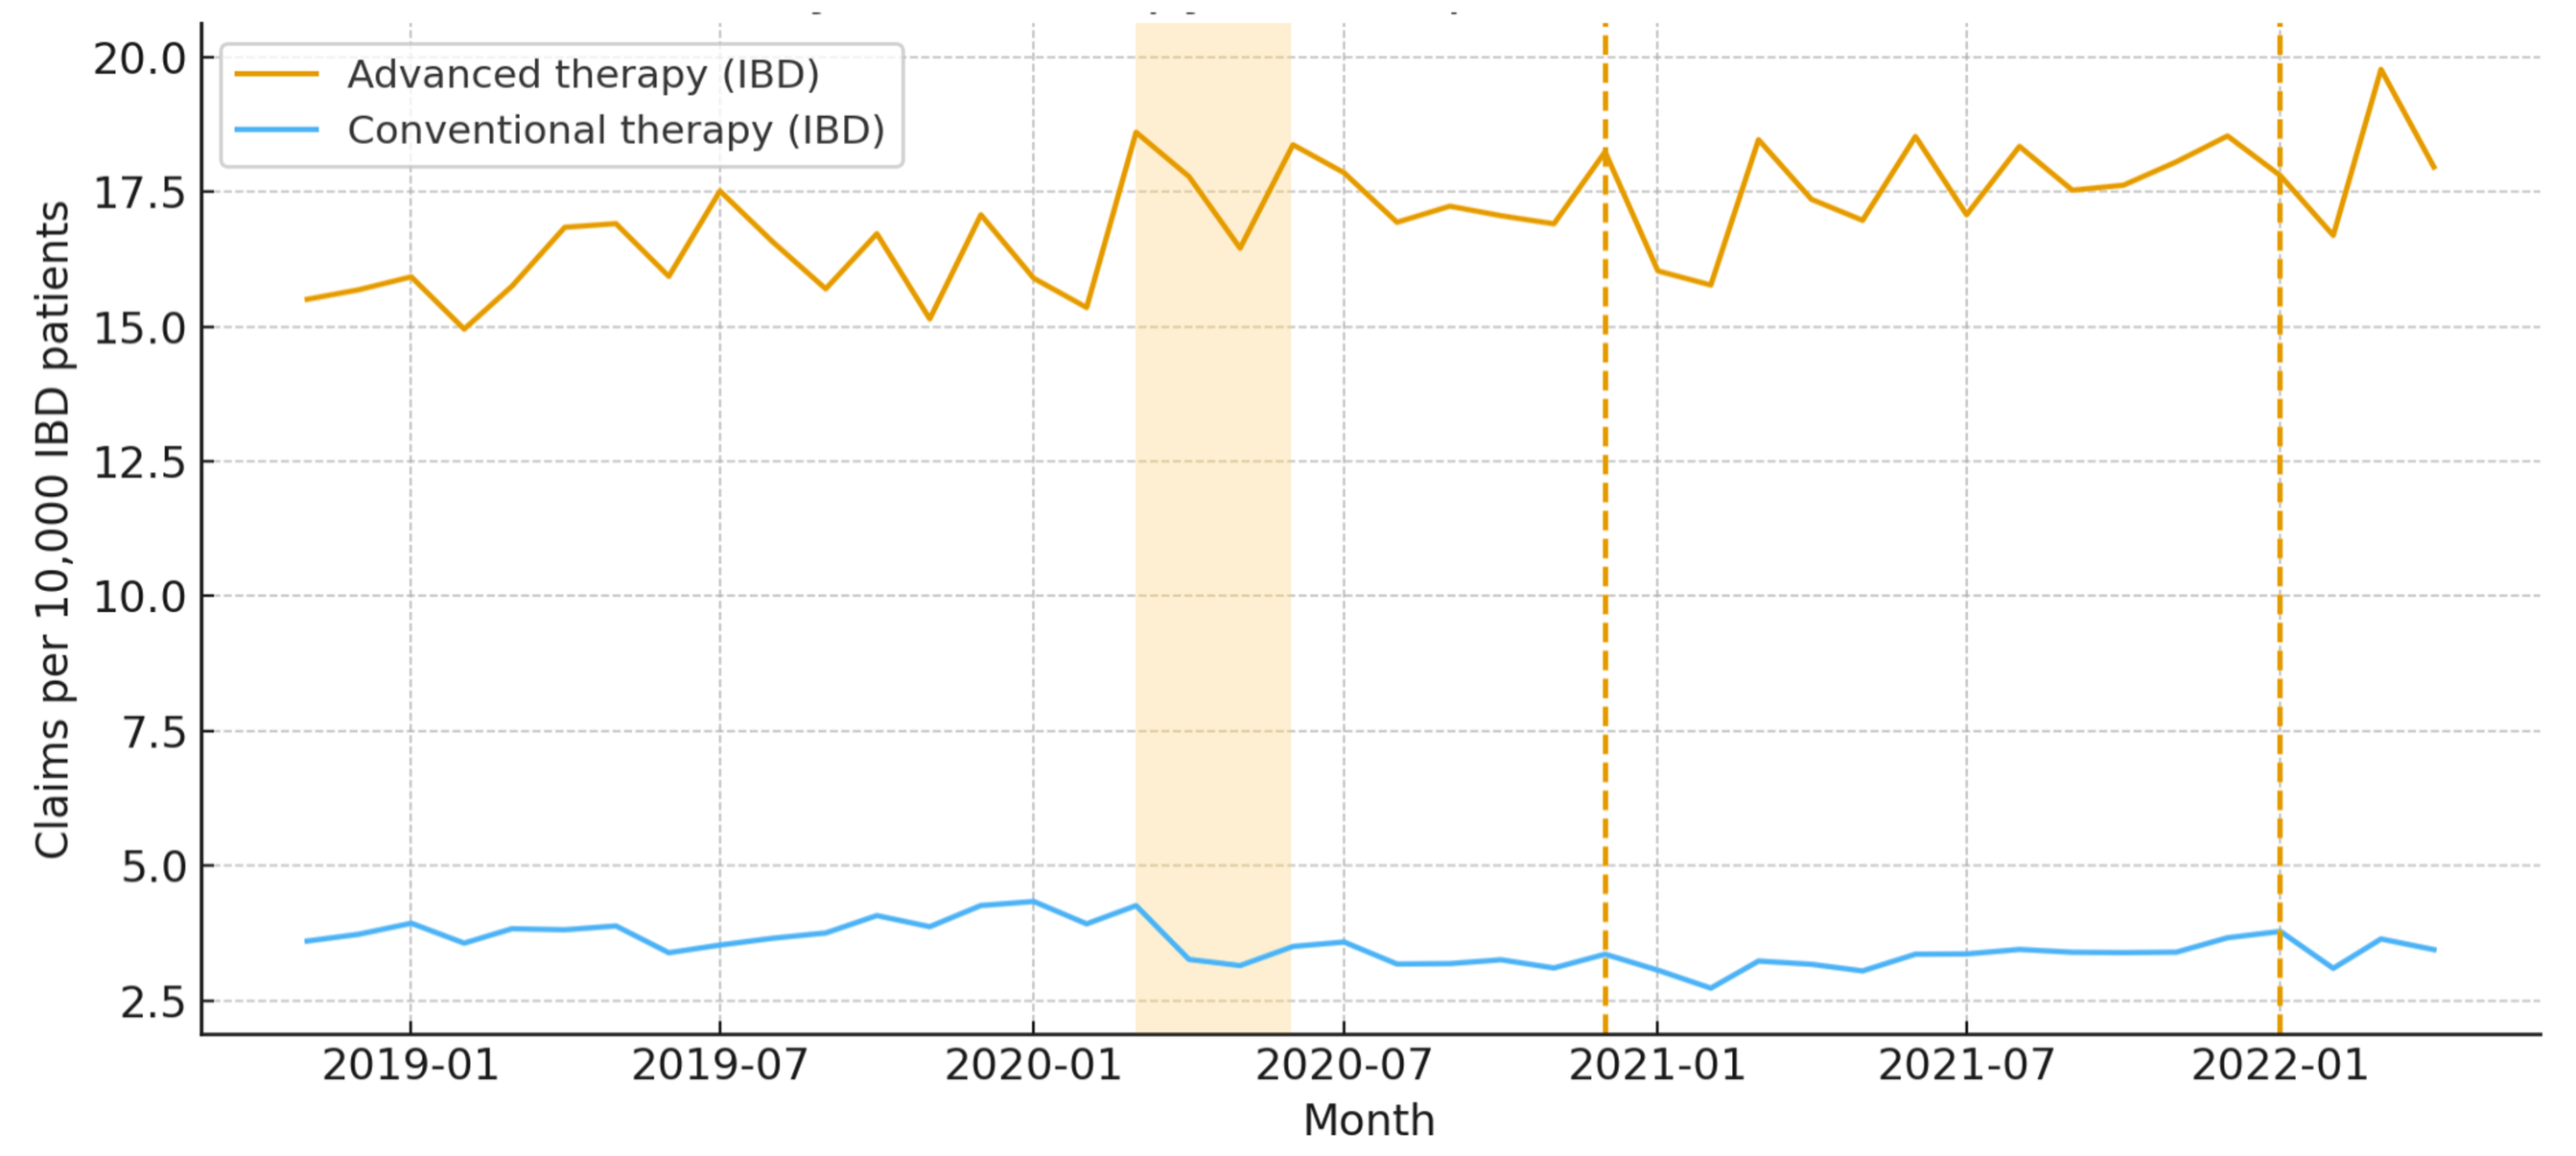
Advanced therapy (IBD)
Conventional therapy (IBD)
Claims per 10,000 IBD patients
Month

Figure 2. Recovery and Evolution of Advanced Therapy Utilization Post-Pandemic Onset.
After a transient decline during early 2020, utilization of advanced IBD therapies rebounded and ultimately exceeded pre-pandemic levels. Analysis of aggregate pharmacy and medical claims (PearlDiver) indicates a therapeutic shift toward self-administered and oral formulations, supporting sustained disease control and treatment continuity under evolving care constraints. Shaded region indicates the early-disruption phase (Jan–Jul 2020); vertical dotted lines denote key recovery milestones (Jan 2021 and Jan 2022).

Relative Growth in Telehealth Utilization During COVID-19

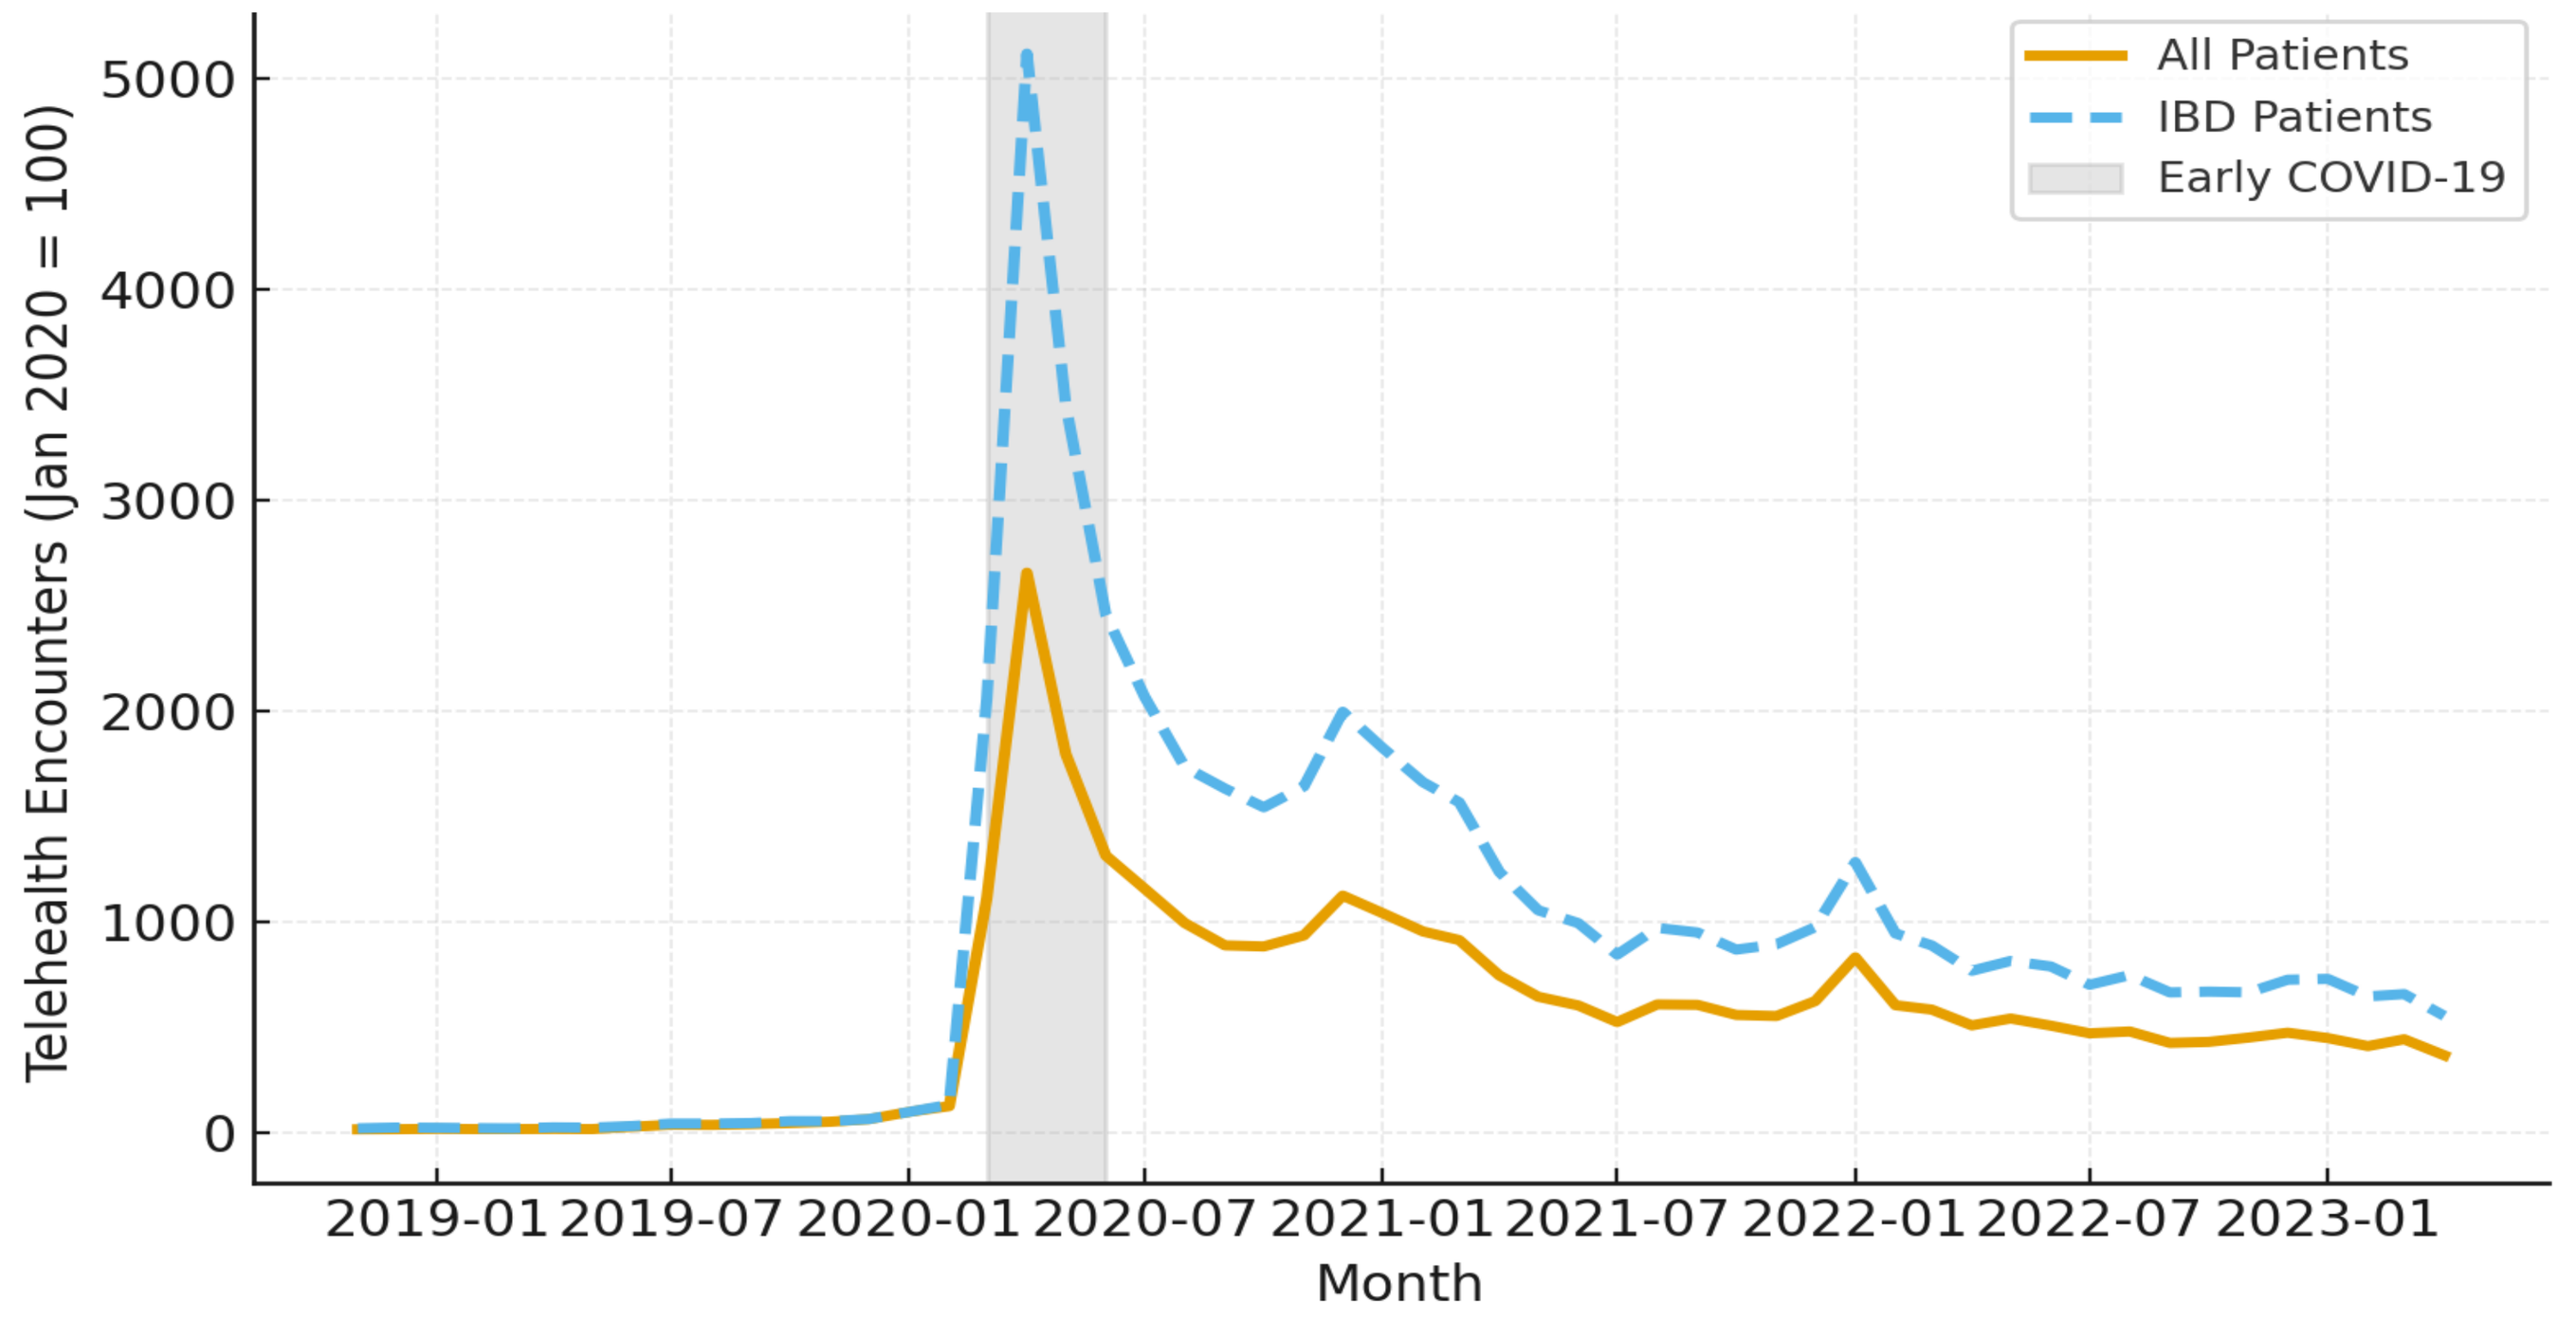
All Patients
IBD Patients
Early COVID-19
Telehealth Encounters (Jan 2020 = 100)
Month

Figure 3. Relative Growth in Telehealth Utilization During the COVID-19 Pandemic.
Monthly telehealth encounters increased sharply during early 2020 (gray shaded region), reflecting the rapid expansion of virtual care as healthcare systems adapted to pandemic-related access restrictions. Utilization remained above pre-pandemic levels through subsequent years, indicating sustained integration of telemedicine into routine practice. Data from PearlDiver Mariner (January 2019–April 2023) for all patients and those with inflammatory bowel disease are presented relative to January 2020 levels (set as 100) to illustrate proportional growth and the sustained role of telehealth in supporting care continuity.

CONCLUSION

- COVID-19–related disruptions led to early reductions in IBD therapy utilization and in-person care.
- IBD vaccination rates initially lagged but reached population levels by late pandemic phases.
- Rapid telehealth expansion supported continuity of care and re-engagement with preventive services during the recovery period.
- Strengthening adaptable care systems is essential for future public health emergencies.

Monthly COVID-19 Vaccinations Per 10,000 Patients

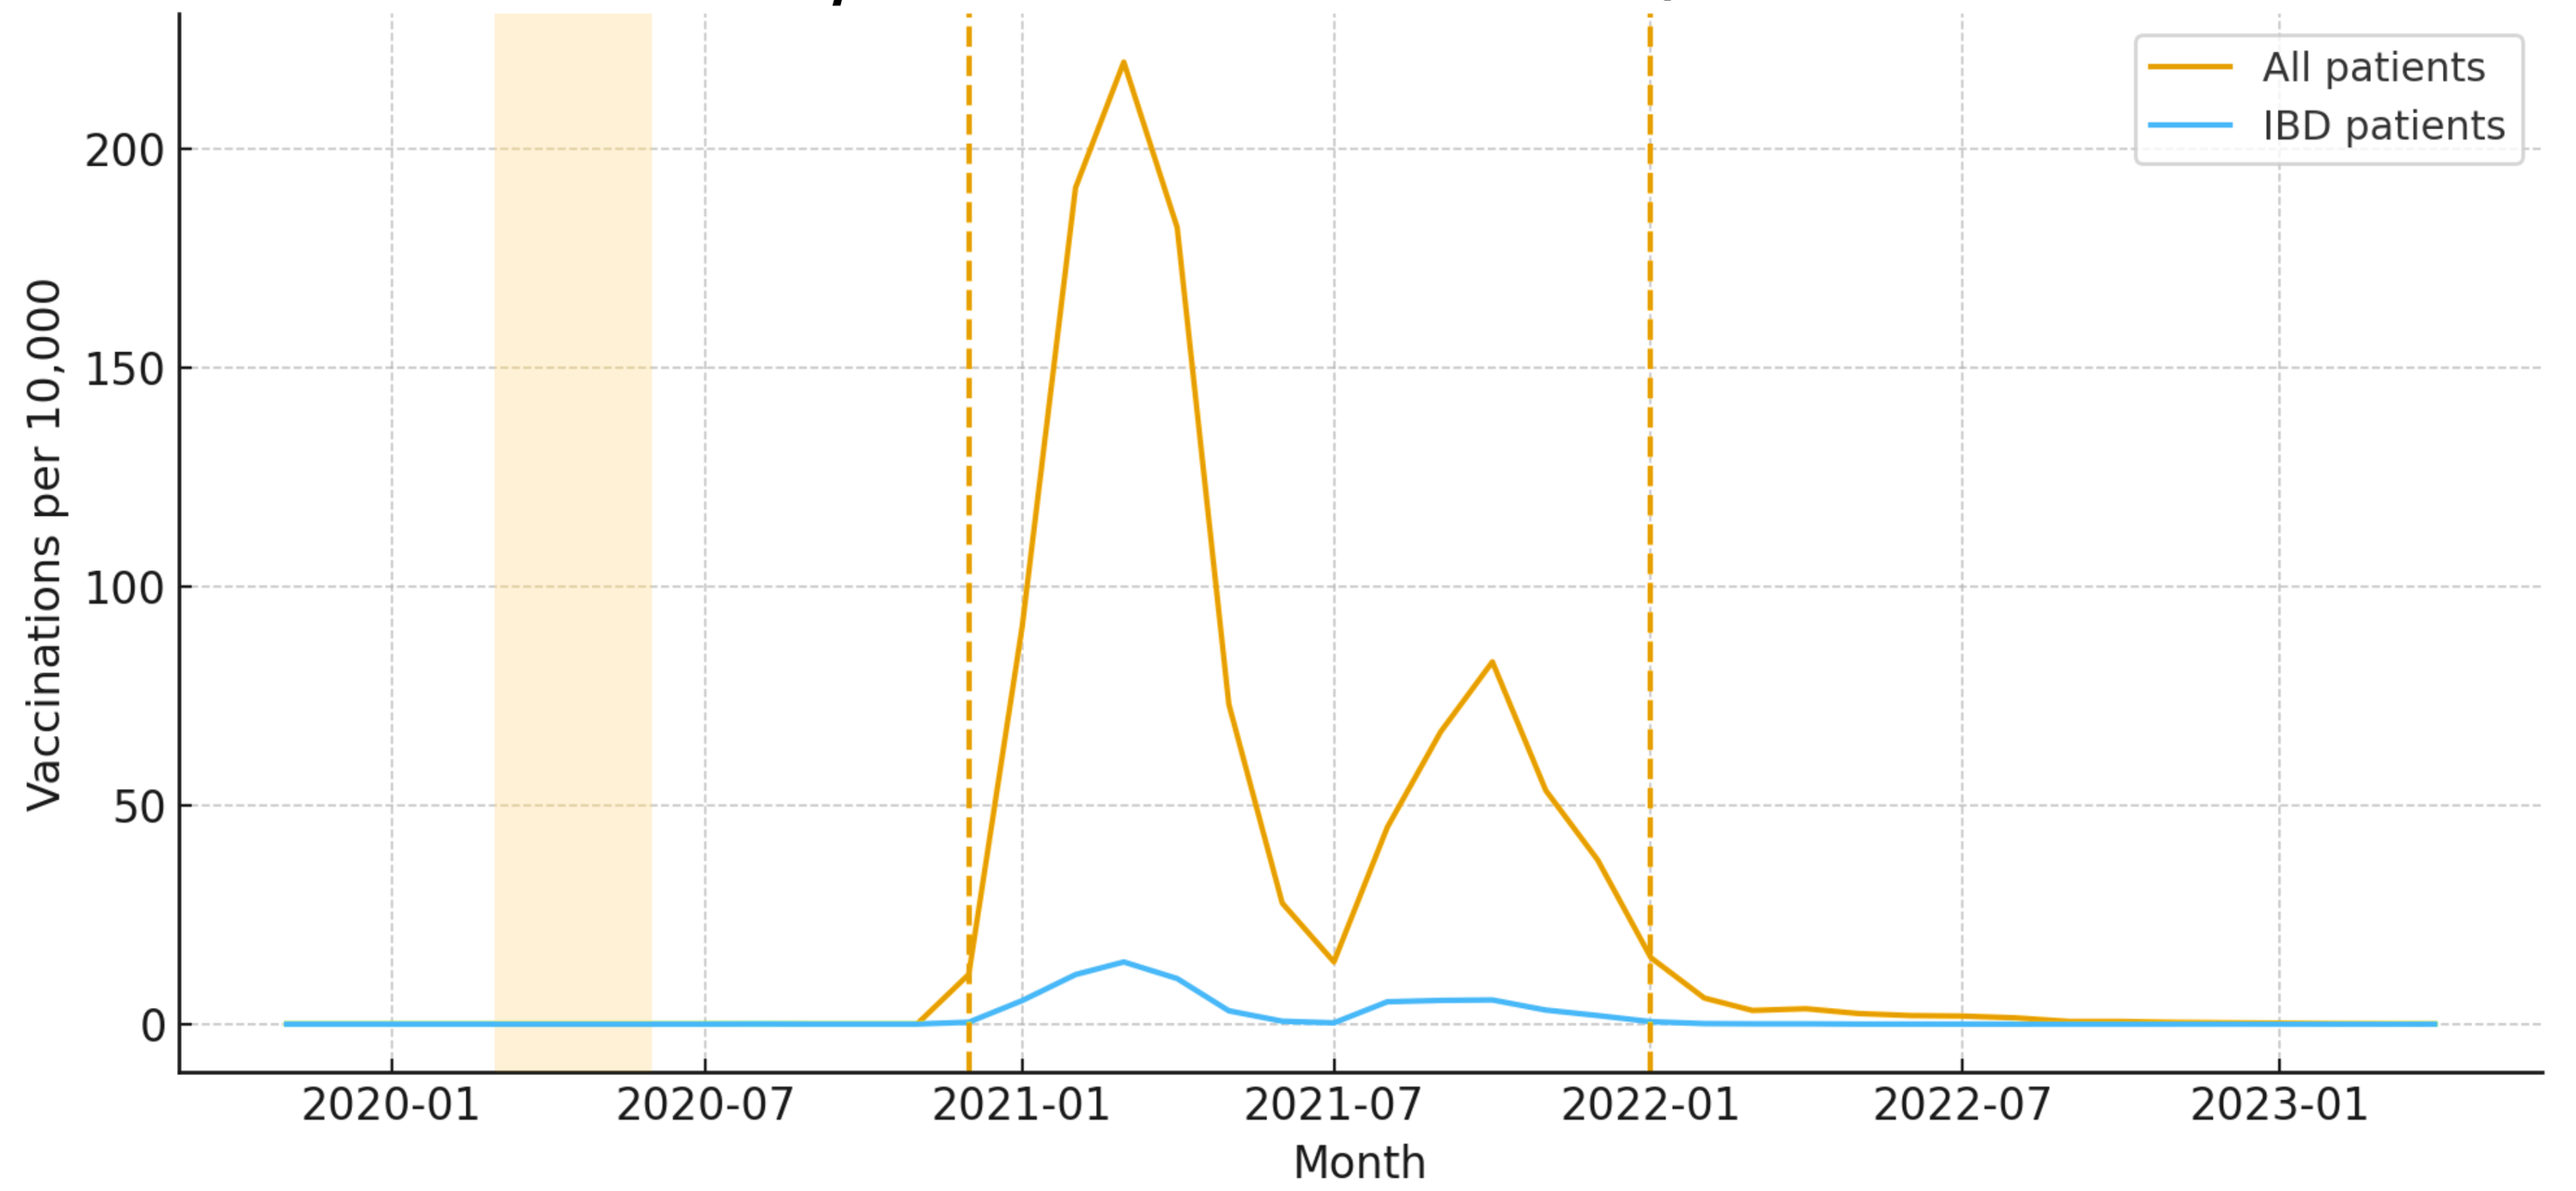
All patients
IBD patients
Vaccinations per 10,000
Month

Figure 4. COVID-19 Vaccination Uptake Among Patients With IBD (2020–2023).
After the December 2020 rollout, vaccination rates among IBD patients increased rapidly and converged toward general U.S. population trends by mid-2021, though with a slightly earlier secondary rise indicating proactive uptake once vaccine safety and accessibility improved. Data from PearlDiver Mariner vaccination claims illustrate re-established preventive care engagement as healthcare access normalized. Shaded region indicates the early-disruption phase; vertical dotted lines show key recovery milestones (Jan 2021 and Jan 2022).

Monthly IBD Hospital Admission Per 10,000 IBD Patients

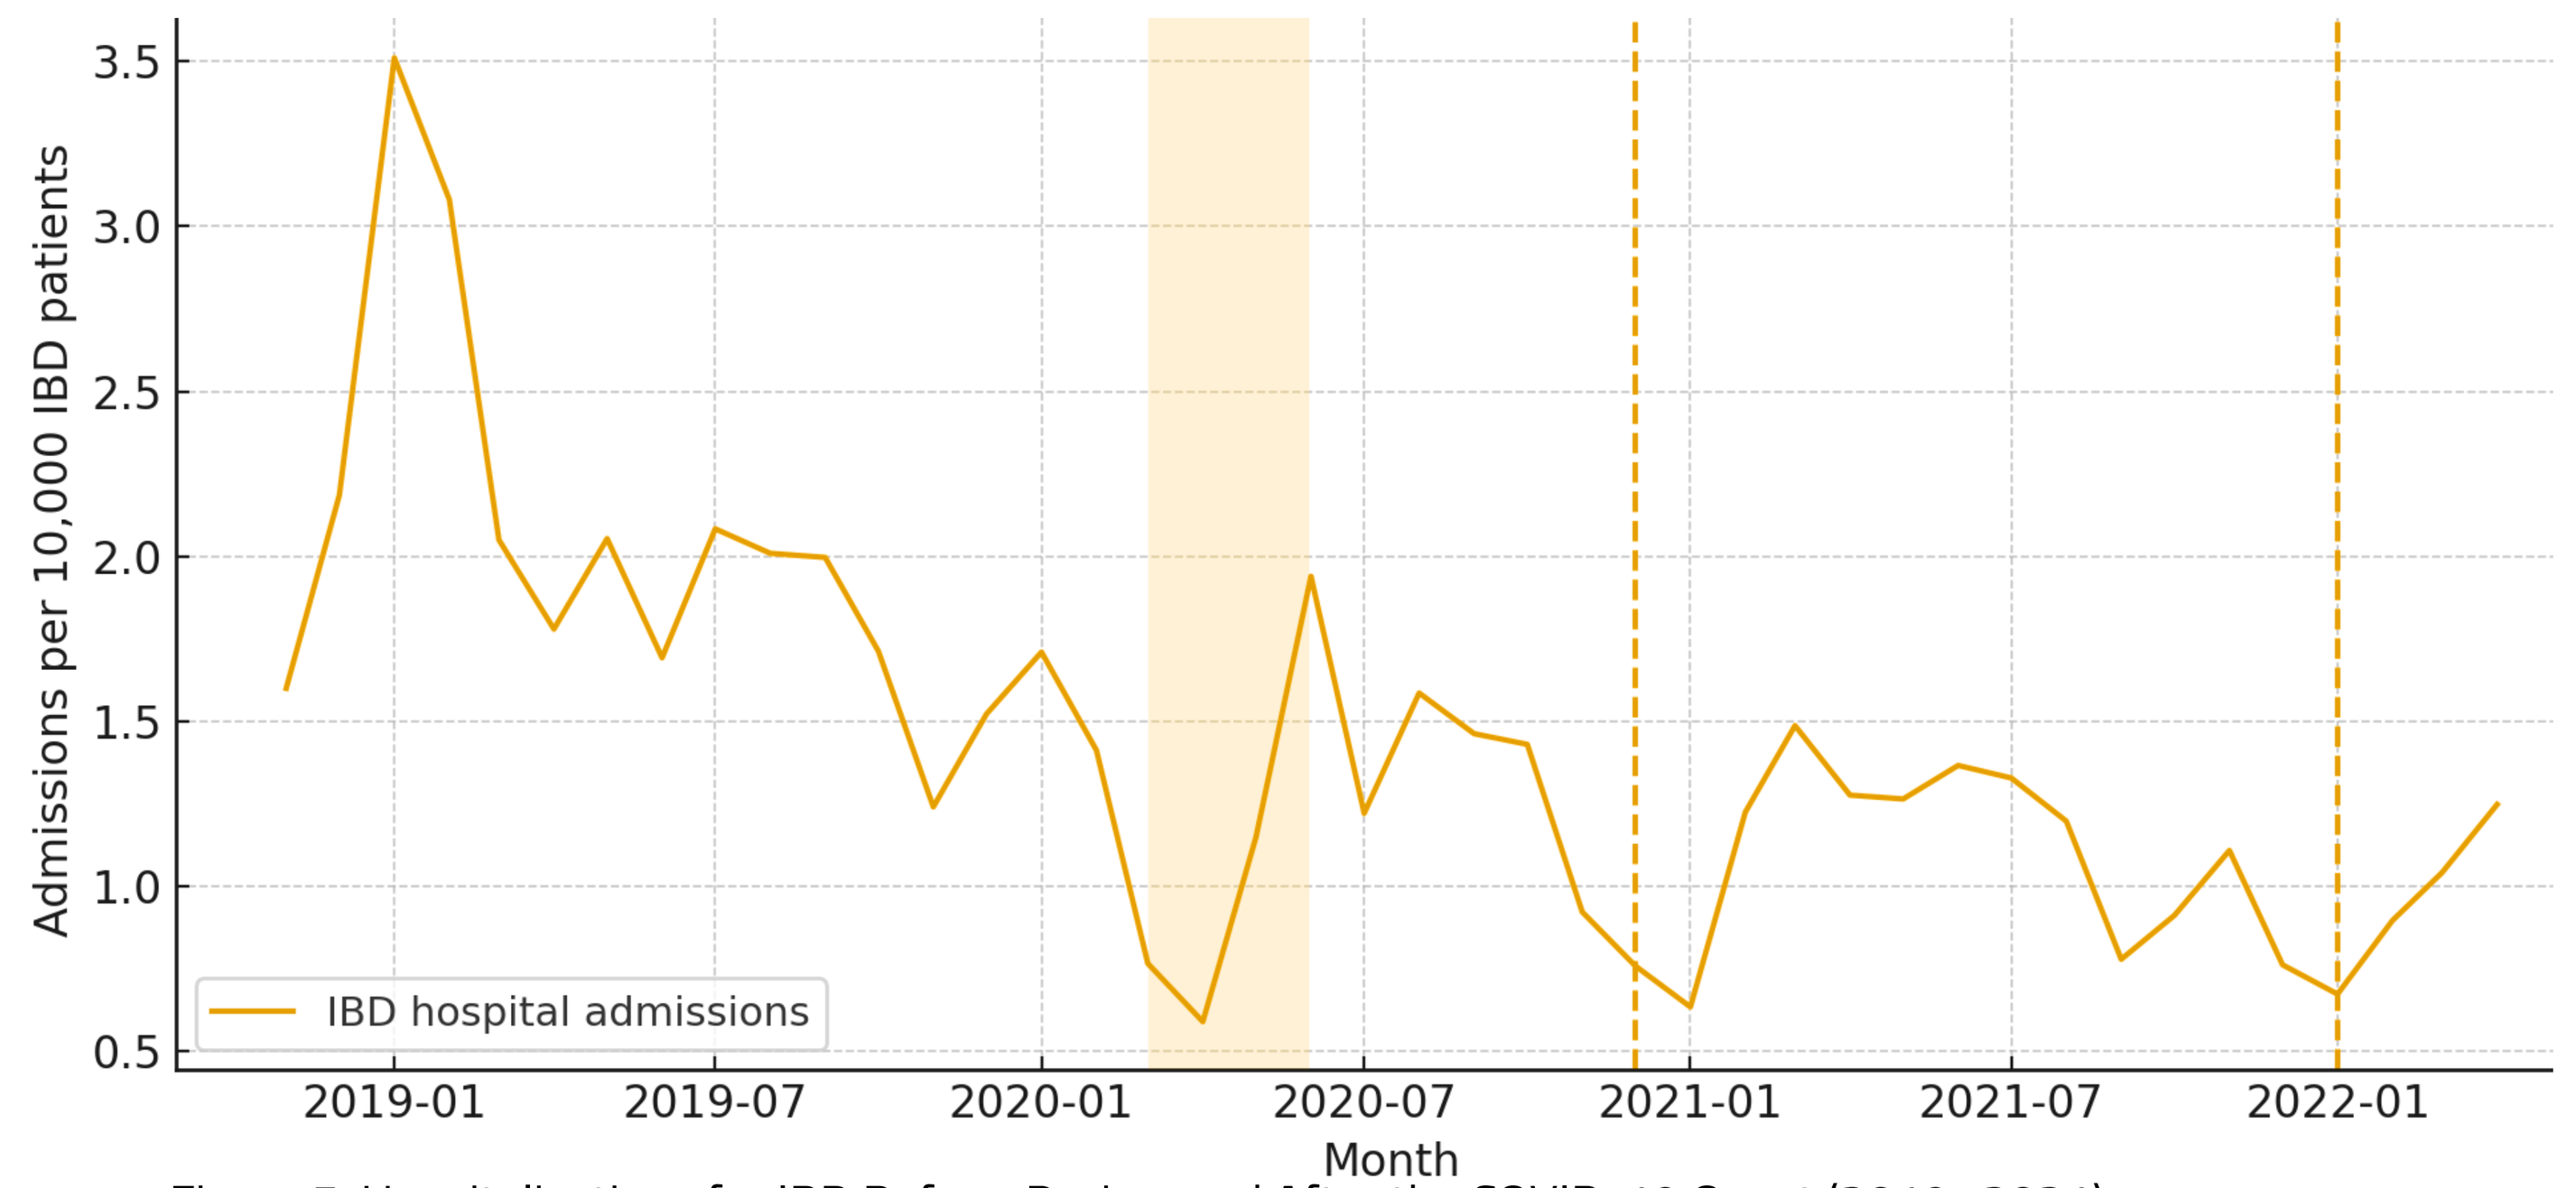
IBD hospital admissions
Admissions per 10,000 IBD patients
Month

Figure 5. Hospitalizations for IBD Before, During, and After the COVID-19 Onset (2019–2021).
IBD-related hospitalizations dropped sharply in March 2020 and remained below pre-pandemic levels throughout 2021. The pattern, derived from national hospitalization claims within the PearlDiver Mariner database, suggests postponed elective procedures and a system-wide shift toward outpatient or telehealth-based management during periods of restricted hospital access. Shaded region indicates the early-disruption phase; vertical dotted lines show key recovery milestones (Jan 2021 and Jan 2022).

METHODOLOGY

- Design: Retrospective cohort study using U.S. administrative claims data (PearlDiver Mariner; November 2019–April 2023).
- Cohorts: IBD patients pre-COVID (Nov 2019–Feb 2020) vs post-COVID (Mar 2020–Apr 2023).
- Analyses: Monthly national trends in IBD-related encounters, therapy utilization, telehealth adoption, vaccination uptake, and hospitalizations were evaluated across four pandemic phases:
 pre-pandemic (Jan–Dec 2019), early disruption (Mar–May 2020), adaptation (Jun–Dec 2020), and recovery (Jan 2021–Jan 2022)
 - Primary: Telehealth encounters, vaccination rates, IBD therapy utilization, and hospitalizations over time.
 - Secondary: Trends assessed by pandemic phase to characterize evolving care delivery and access patterns

RECOMMENDATIONS

- Strengthen telehealth infrastructure and flexible treatment delivery models (e.g., home infusions, self-administered or oral therapies) to maintain continuity of care during healthcare disruptions.
- Establish contingency protocols and proactive provider–patient communication frameworks to sustain IBD management during future public health emergencies
- Enhance vaccination outreach and align clinical practice with AGA/ACG guidance to protect immunosuppressed patients and improve preventive care uptake.
- Implement real-time data monitoring and regional surveillance systems to rapidly identify care gaps and guide resource allocation during emerging crises.
- Address access and equity disparities revealed in early-pandemic data, ensuring consistent and equitable IBD care across regions most affected by service disruptions.

REFERENCES

1. Lin S, Lau LH, Chanchlani N, Kennedy NA, Ng SC. Recent advances in clinical practice: management of inflammatory bowel disease during the COVID-19 pandemic. Gut. 2022;71(7):1426-1439. doi:10.1136/gutjnl-2021-326784
2. El Ouali S, Rubin DT, Cohen BL, Regueiro MD, Rieder F. Optimal inflammatory bowel disease management during the global coronavirus disease 2019 pandemic. Curr Opin Gastroenterol. 2021;37(4):313-319. doi:10.1097/MOG.0000000000000741
3. Rubin DT, Feuerstein JD, Wang AY, Cohen RD. AGA clinical practice update on management of inflammatory bowel disease during the COVID-19 pandemic: expert commentary. Gastroenterology. 2020;159(1):350-357. doi:10.1053/j.gastro.2020.04.012
4. Viganò C, Mulinacci G, Palermo A, et al. Impact of COVID-19 on inflammatory bowel disease practice and perspectives for the future. World J Gastroenterol. 2021;27(33):5520-5535. doi:10.3748/wjg.v27.i33.5520
5. Esposito S, Caminiti C, Giordano R, et al. Risks of SARS-CoV-2 infection and immune response to COVID-19 vaccines in patients with inflammatory bowel disease: current evidence. Front Immunol. 2022;13:933774. doi:10.3389/fimmu.2022.933774
6. Farraye FA, Melmed GY, Lichtenstein GR, et al. ACG clinical guideline update: preventive care in inflammatory bowel disease. Am J Gastroenterol. 2025;120(7):1447-1473. doi:10.14309/ajg.0000000000003541